Potential health and safety risks to employees and environment from cell sorting operations and possible aerosolization or other exposure incidents that could potentially contaminate other operations in the laboratory

Youngmi A. Girard, COHN-S, MBA[1]; Victor D'Amato, CIH , CSP[2]; and Kimberly DiGiandomenico, MS, RBP[3]

[1]Safety, Health and Environment, MedImmune, LLC ; [2,3] Safety Consultant, Atrium Environmental Health & Safety Services, LLC

Abstract

In the National Institutes of Health (NIH) Biosafety Policy for Cell Sorters, cell sorters are defined as instruments that perform analysis of cells or particles, and that isolate desired cells into droplets that are deflected electrostatically into open collection vessels. Cell sorters are known as stream-in-air or jet-in-air cell sorters, which describes the ejection of fluid from the nozzle into the open air. The likelihood of aerosol production by cell sorters is relatively high due to the possibility of fluid exiting a small orifice at high pressures impacting a hard surface. The risk of aerosol production is highest if a partial obstruction of the nozzle orifice and subsequent stream deviation occurs.

The objective of this assessment was to determine potential health and safety risks to employees and the environment from the MedImmune, LLC cell sorting operations and to identify possible aerosolization or other exposure incidents that could potentially contaminate other operations in the laboratory. To determine the appropriate laboratory containment level suitable for the cell sorting operations conducted at MedImmune, LLC, this study assessed the following areas through Qualitative Aerosol Deposition testing:

- The laboratory space, including general housekeeping practices as they relate to operator health and safety and potential cross-contamination;
- Operation of the BD FACS Aria II including engineering controls for minimizing operator exposures and cross-contamination; and
- Operator practices and procedures established to minimize operator exposures and cross-contamination.

Materials & Methods

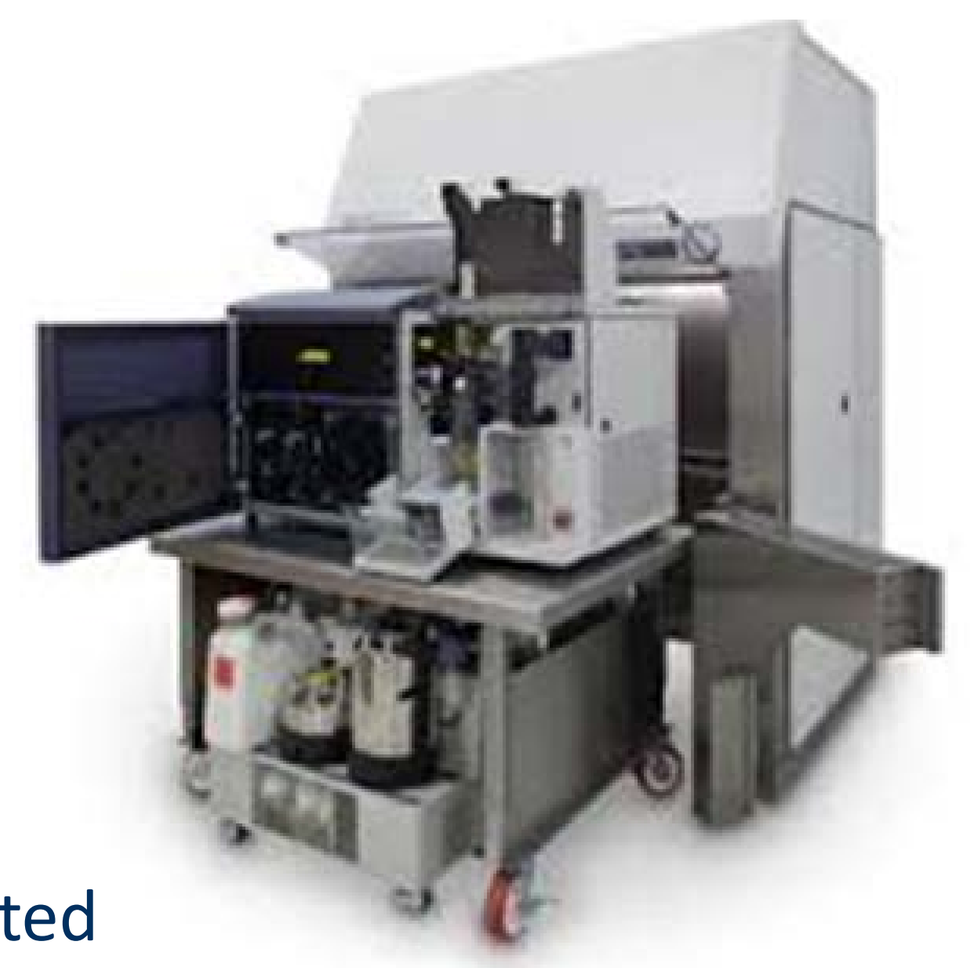

The objective of this assessment was to focus on the extent of possible fugitive emissions and the distance they travel from the BD FACS Aria II contained within a Baker BioProtect biological safety cabinet (BSC). Therefore, it was determined that an easily implemented, safe, qualitative method for assessing aerosol deposition should be conducted.

Qualitative aerosol deposition testing was conducted to verify aerosol containment though the use of the integrated BSC on the BD FACS Aria II. The test plan was based on a similar study referenced by Holmes in which an ultraviolet (UV)-excitable dye (Clear Blue Fluorescent Water Tracer Dye (CBD); Risk Reactor, Santa Ana, CA, Order Code IFWB-C0) was used to distinguish aerosols originating from the cell sorter from ambient particles. The CBD tracer is "excitable" and fluoresces bright blue at a UV wavelength of 365 nanometers (nm). The CBD was loaded into the sample tube. Paper fluorescence was measured using a hand-held 365nm UV lamp. Filters showing any visual fluorescence were considered "contaminated." In addition, the operator's gloves and lab coat were tested for fluorescence following each test event.

Test Methodology

***Equipment and Materials*:**

- 50 mL water-based fluorescent dye, excitable at a UV wavelength of 365 nm
- UV lamp operating at 365nm
- Dark box with openings for viewing and UV lamp
- 100 paper filters, P4 Grade, 7cm diameter
- Nitrile gloves
- Cell sorter sample tubes and collection plate
- 200 µL pipette with disposable tips
- Small cotton-tip swab (to simulate obstruction)
- Digital camera

Optimal Dye Concentration was determined by evaluating the concentration at which fluorescent aerosols could be identified using a hand-held 365nm UV lamp. Based on the flow rate of the sheath solution for the FACS Aria II, it was determined that the final dye concentration at the plate was 1:320.

Filter Placement: Filters were placed on horizontal surfaces at various locations throughout the laboratory, as well as on and around the BD FACS Aria II (Figure 3).

***Test Events*:**
Each of the following operating scenarios of the BD FACS Aria II were tested using a 100 µm nozzle at 27 psi nozzle pressure. For Tests A & B, 200uL of dye in the sample tube for a 5 minute sort run was used, and for Test C, 200mL of dye in the sample tube with a 1 minute sort run was used. *Note: The BSC remained 'on' during all scenarios*:

A. Normal operating mode with collection chamber and BSC shield closed.
B. Normal operating mode with collection chamber and BSC shield open.
C. Failure mode (simulated an obstructed nozzle) with collection chamber and BSC shield open.

Results

Test A: Normal operating mode with the collection chamber and BSC shield closed and Test B: Normal operating mode with collection chamber and BSC shield open. All collection sites were negative (1) with the exception of next to and on top of the collection site (2 and 3 (Test A and Test B, respectively)

Figure 1: Representative negative (1) and positive samples (2, 3)

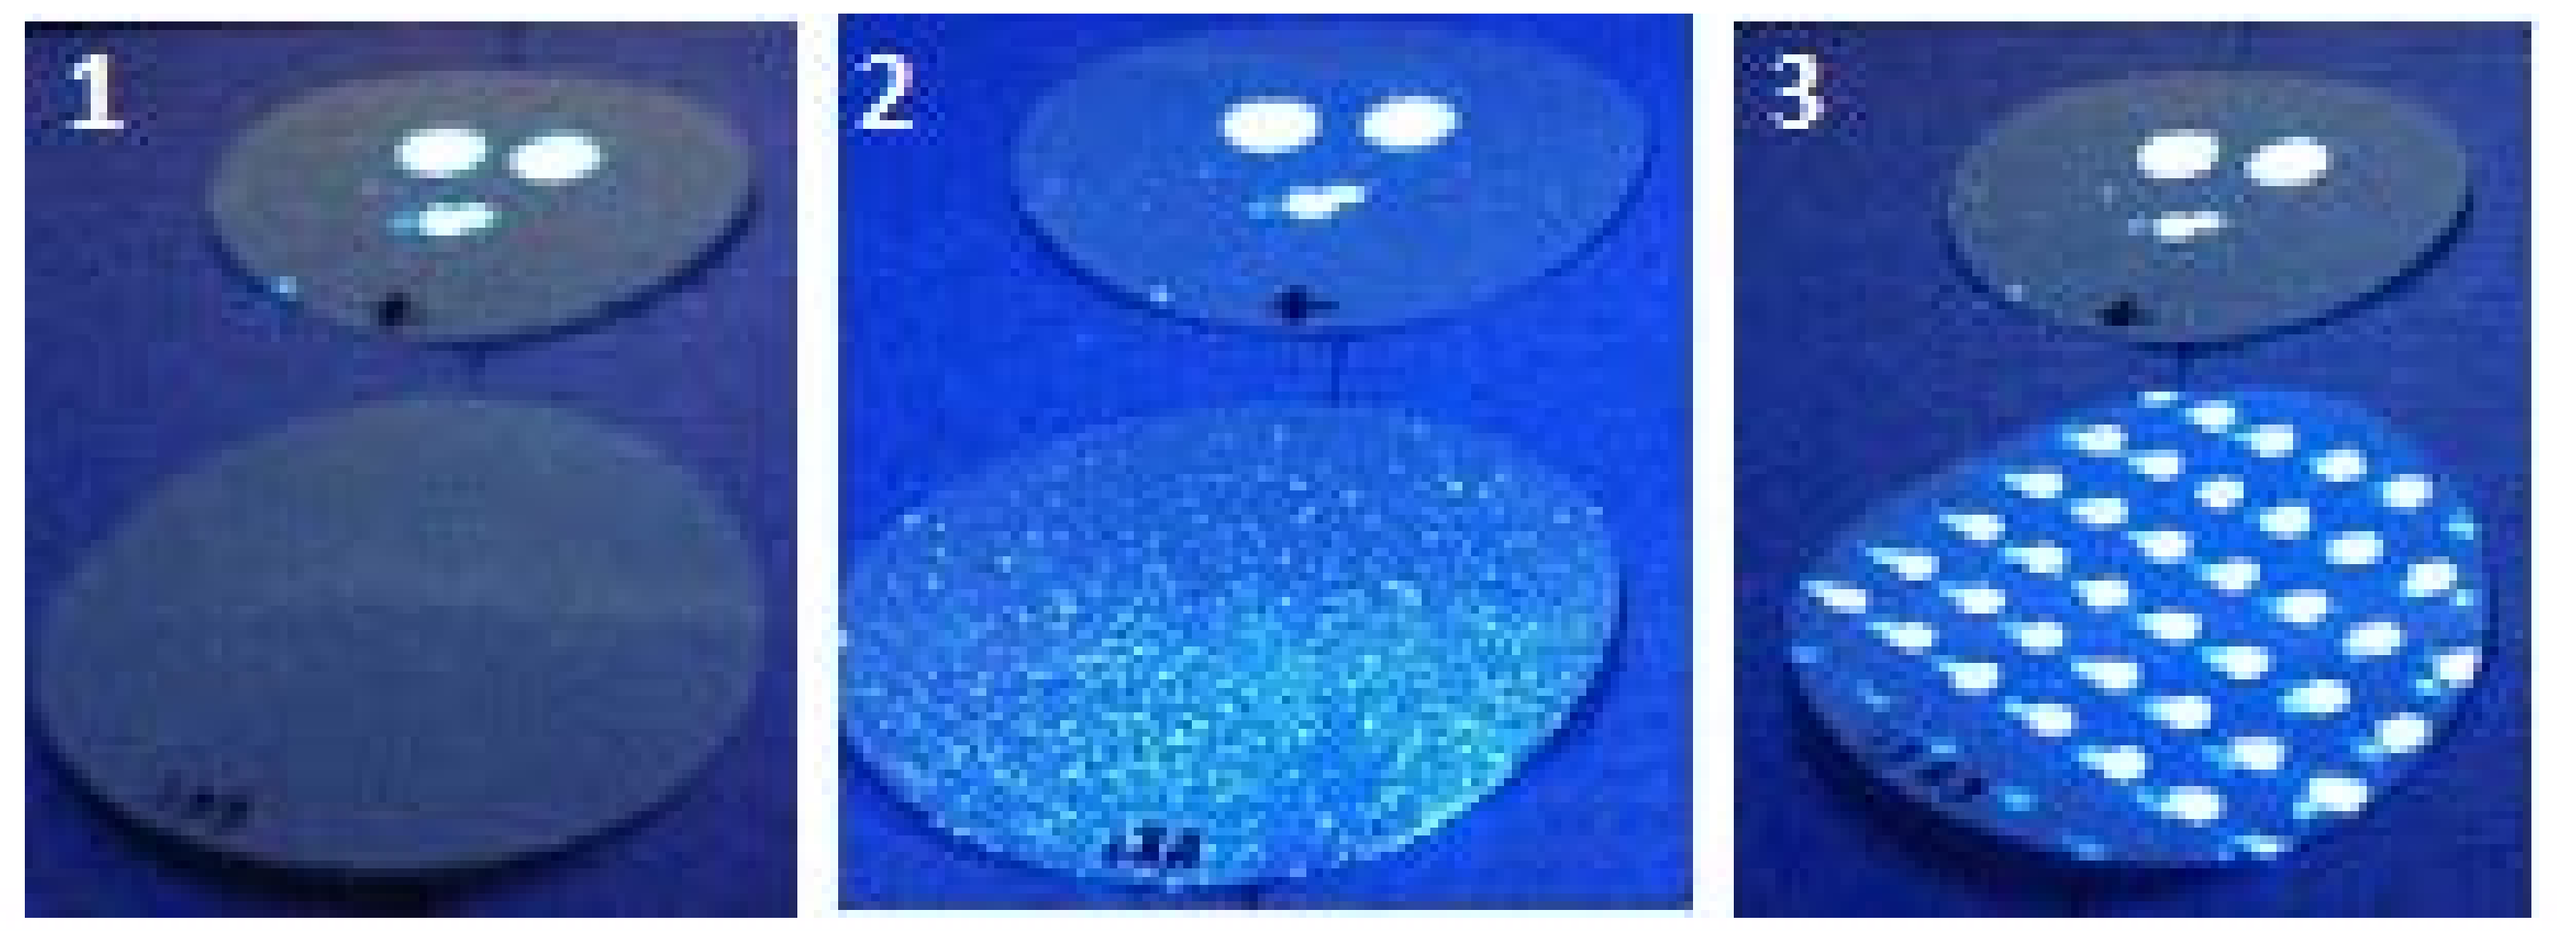
1
2
3

Test C: Simulated obstruction in the cell sorting stream with the collection chamber and BSC shield open. All collection sites were negative with the exception of next to and on top of the collection site.

Figure 2: Test C: Simulated obstruction and fluorescent visualization of obstruction

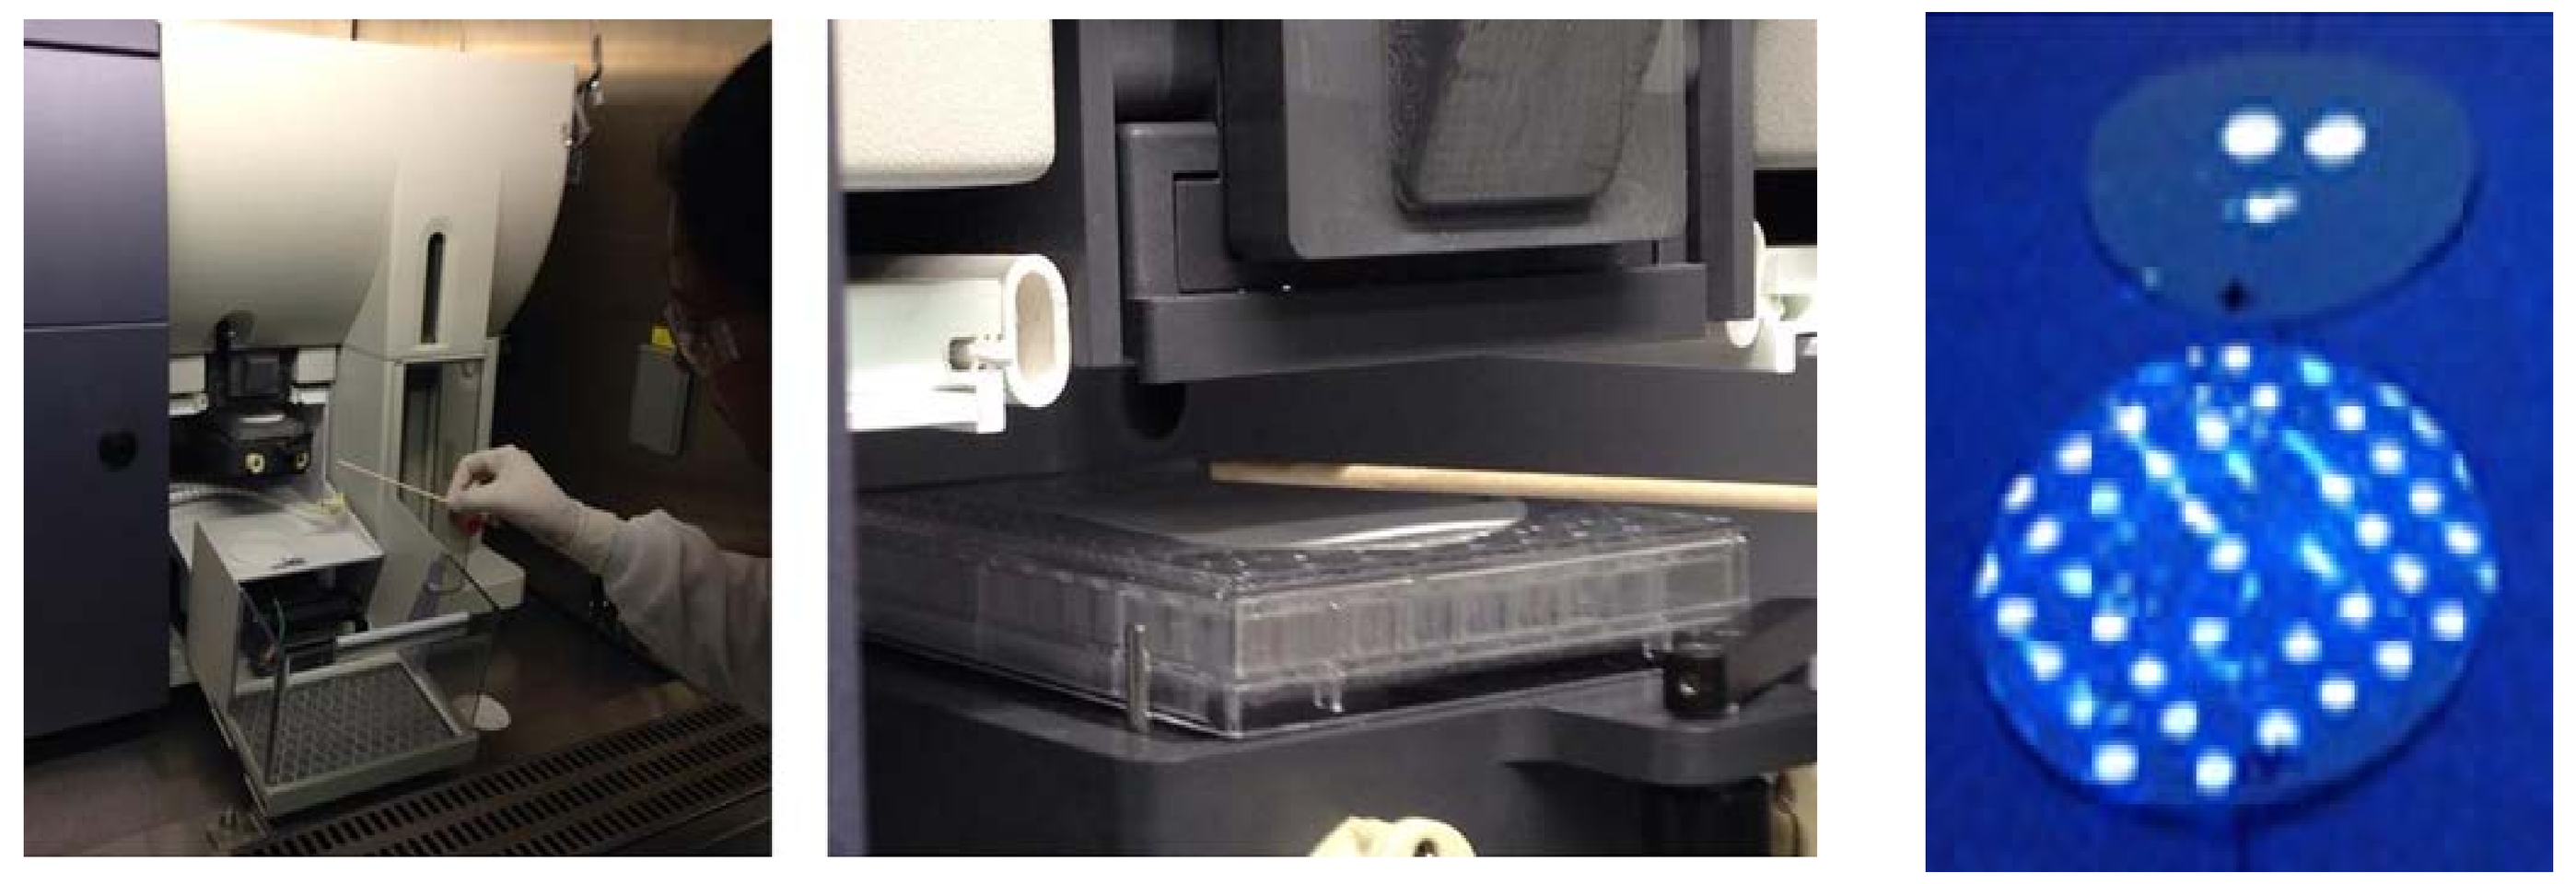

Figure 3: Test events A, B, and C and resulting observations

Test Event	A		B		C	
Description	Normal Sorting Operation, Chamber Closed		Normal Sorting Operation, Chamber Open/BSC Open		Stream Disruption (Simulated Obstruction), Chamber Open/ BSC Open	
Results by Location	pos/neg	density	pos/neg	density	pos/neg	density
Operator gloves (before)	NEG	N/A	NEG	N/A	NEG	N/A
Operator lab coat (before)	NEG	N/A	NEG	N/A	NEG	N/A
1. Inside FACS enclosure, on surface in front of the collection chamber	NEG	N/A	NEG	N/A	NEG	N/A
2. Inside BSC below AMS closed connection	NEG	N/A	NEG	N/A	NEG	N/A
3. At FACS BSC exhaust	NEG	N/A	NEG	N/A	NEG	N/A
4. On FACS computer module keyboard	NEG	N/A	NEG	N/A	NEG	N/A
5. On FACS computer module mouse	NEG	N/A	NEG	N/A	NEG	N/A
6. Across from BSC	NEG	N/A	NEG	N/A	NEG	N/A
7. On bench parallel to #6 above in next bay	NEG	N/A	NEG	N/A	NEG	N/A
8. On table by LSRII-Green	NEG	N/A	NEG	N/A	NEG	N/A
9.On ISXOL-AMNIS keyboard (on bench behind BSC)	NEG	N/A	NEG	N/A	NEG	N/A
10. Keyboard of MACSQUANT-03	NEG	N/A	NEG	N/A	NEG	N/A
11. On Bench by FACSCALIBUR-01	NEG	N/A	NEG	N/A	NEG	N/A
12. FACS Inside sorting chamber	POS	12 droplets	POS	6 droplets	POS	**7 droplets**
13. FACS on collection plate (positive control)	**POS**	**70% coverage**	**POS**	**100% coverage**	**POS**	**100% coverage**
Operator gloves (after)	NEG	N/A	NEG	N/A	NEG	N/A
Operator lab coat (after)	NEG	N/A	NEG	N/A	NEG	N/A

Conclusions

The NIH Biosafety Policy for Cell Sorters and the International Association of Analytical Cytology (ISAC) biosafety standards discuss increasing the biosafety level of agents handled on cell sorters due to the potential for aerosolization. Under these guidance documents, low risk human pathogens that are designated as Risk Group 2 agents handled under normal Biosafety Level 2 (BSL2) laboratory procedures and practices may be re-classified as requiring BSL2 laboratory procedures with enhanced precautions because of the potential for aerosol and/or splash exposure associated with cell sorting. Cell sorting is considered a laboratory procedure hazard under these guidance documents.

Regarding the studies conducted on the BD FACS Aria II at the MedImmune, LLC Gaithersburg facility, no aerosol deposition was measured on surfaces outside the BD FACS Aria II contained within the Baker BioProtect or on the operator's gloved hands or laboratory coat. It was concluded that for the samples being routinely sorted on the contained BD FACS Aria II, BSL2 practices would be sufficient; however, access to the area during a sort would be limited; and the operator was advised to don respiratory protection as an added precaution during loading and unloading of the sort chamber and if clearing a nozzle obstruction, since the greatest risk for exposure would be present during these steps.

It is important to understand that the testing described here was conducted using a 100 µm nozzle operated with a sheath pressure of 27 psi. While these characteristics are representative of most sorting procedures on the BD FACS Aria II, conducting sorts using a different nozzle size and operating at a different sheath pressure may produce different results.

Acknowledgements

Special thanks to Chris Groves, Sr. Manager-Respiratory Inflammation and Autoimmunity (RIA); Radhika Rayanki, R&D Associate II-RIA; and Jeff Carrell , Scientist I-RIA for use of their equipment as well as time and expertise to conduct these experiments.

References

BD FACSAria II User's Guide
NIH Biosafety Policy for Cell Sorters, July 30, 2012
International Society for the Advancement of Cytometry Cell Sorter Biosafety Standards, Holmes, K.L., et. al., *Cytometry Part A* 85A: 434-453, 2014
Characterization of Aerosols Produced by Cell Sorters and Evaluation of Containment, Holmes, K. L., *Cytometry A*. 2011, December; 79(12): 1000–1008
Novel Rapid Method for Visualization of Extent and Location of Aerosol Contamination During High-Speed Sorting of Potentially Biohazardous Samples, Oberyszyn, A.S., Robertson, F.M., *Cytometry*, 43:217–222 (2001).
Testing the Efficiency of Aerosol Containment During Cell Sorting, Schmid, I., Hultin, L.E., Ferbas, J., *Current Protocols in Cytometry* (1997) 3.3.1-3.3.15.
A Method to Assess Leakage from Aerosol Containment Systems: Testing a Fluorescence-Activated Cell Sorter (FACS) Containment System Using the Radionuclide Technetium-99m, Wallace, R.G., Aguila, H.L., Fomenko,J., Price, K.W., *Applied Biosafety,* Vol. 15, No. 2, 2010
Experimental Study of Dispersion and Deposition of Expiratory Aerosols in Aircraft Cabins and Impact on Infectious Disease Transmission, G. N. Sze To, M. P. Wan, C. Y. H. Chao, L. Fang, A. Melikov, *Aerosol Science and Technology*, Vol. 43, Iss. 5, 2009

Poster presented at the American Biological Safety Association 57th Annual Biological Safety Conference in San Diego, CA; October 3-8, 2014